ASCO 2010
Abstract # 6534

Thymic renewal and anti-leukemic effect in adults after haploidentical transplantation and suicide gene therapy

C. Bordignon[1], L. Vago[2], G. Oliveira[2], D. Ghio[2], M.T. Lupo Stanghellini[2], J. Peccatori[2], A. Bondanza[2], A. Lambiase[1], F. Ciceri[2], C. Bonini[2]

[1]Molmed, Milan, Italy; [2]Istituto Scientifico San Raffaele, Milan, Italy

Background

- Haematopoietic stem cell transplantation from partially HLA-matched family donors (haplo-HSCT) represents a promising therapy for high-risk leukaemia
- The infusion of donor T cells transduced with the HSV-TK suicide gene (TK cells) provides a rapid immune-reconstitution while controlling Graft-versus-Host-Disease (GvHD)
- Here we investigated the contribution of TK cells to post-transplantation thymic renewal and Graft-versus-Leukaemia (GvL) effect

Figure 1. TK cell therapy in haploidentical-HSCT

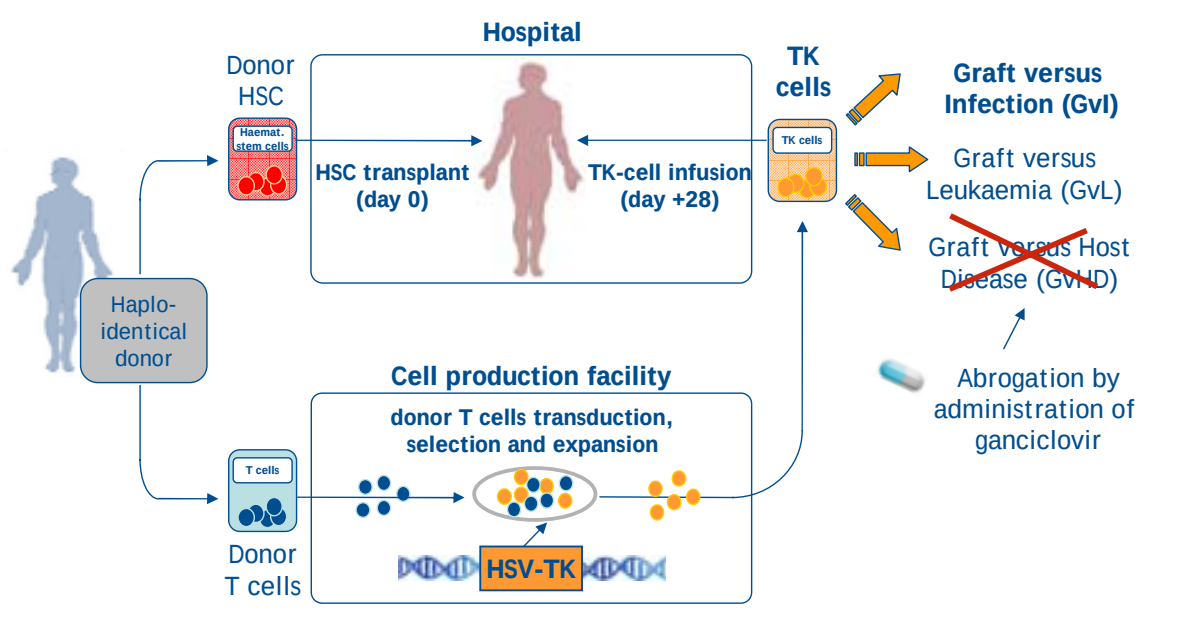

Hospital
Donor HSC
Haemat. stem cells
HSC transplant (day 0)
TK-cell infusion (day +28)
TK cells
Graft versus Infection (GvI)
Graft versus Leukaemia (GvL)
Graft versus Host Disease (GvHD)
Abrogation by administration of ganciclovir
Haplo-identical donor
Cell production facility
donor T cells transduction, selection and expansion
T cells
Donor T cells
HSV-TK

- Donor T cells transduced with the Herpes Simplex Virus Thymidine Kinase gene (TK cells) are selected and expanded *in vitro*
- After 20-40 days from haploidentical HSCT, TK-cells are infused into the patient

Methods

- In a Phase II trial (TK007) of haplo-HSCT for high-risk leukaemia, 28 patients received TK cells (TK+ donor T cells)
- In a selected subset of patients, thymic function was assessed by quantitation of CD31+ in CD4+ naïve T cells, and was correlated with thymic volume, assessed by CT scans
- Alloreactivity against leukemic blasts was studied by MLC

Figure 2. Phase II trial TK007: T cell reconstitution and transplant-related mortality

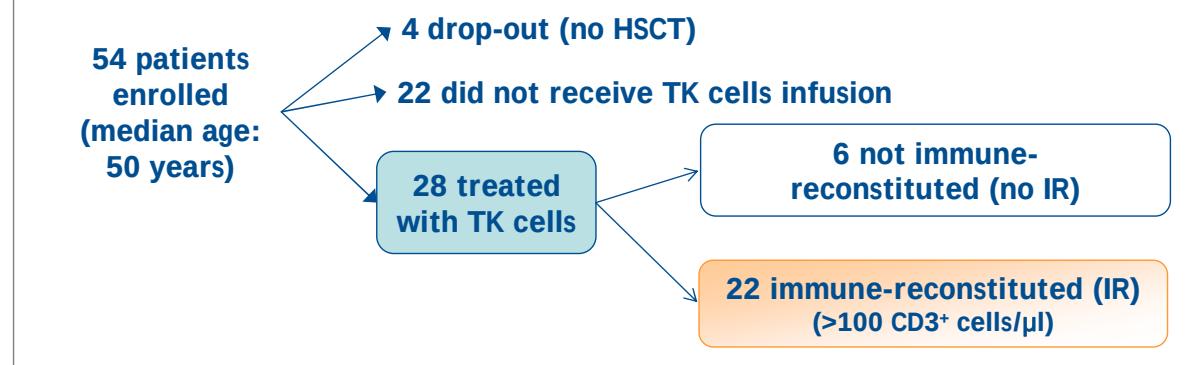

54 patients enrolled (median age: 50 years)
4 drop-out (no HSCT)
22 did not receive TK cells infusion
28 treated with TK cells
6 not immune-reconstituted (no IR)
22 immune-reconstituted (IR) (>100 CD3+ cells/μl)

T cell reconstitution

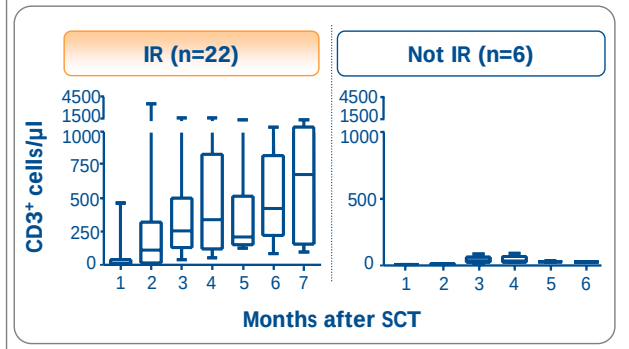

IR (n=22)
Not IR (n=6)
CD3+ cells/μl
Months after SCT

- Patients with TK-cell engraftment promptly achieved CD3+ counts of 100 cells per μl or more, at median time of 75 days from transplant (range 34-127) and 23 days from TK-cell infusion (range 13-42)
- Patients without TK-cell engraftment remained immuno-deficient

Transplant-related mortality

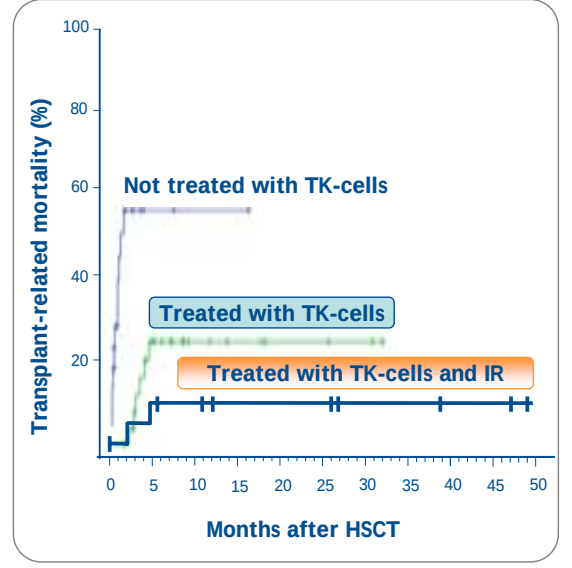

Transplant-related mortality (%)
Not treated with TK-cells
Treated with TK-cells
Treated with TK-cells and IR
Months after HSCT

- TK cells protect from transplant-related mortality

Results

- Post-transplant recovery of TK-negative naïve T cells occurred, reaching values comparable to controls in approximately one year
- After infusion of TK cells , almost all circulating CD4+ naïve T cells were CD31+ Recent Thymic Emigrants, suggesting a direct role of the infused TK cells in promoting thymopoiesis. Accordingly, CT scans documented an increase of thymic volume
- TK cells displayed also a direct anti-leukemic effect, as demonstrated by their selective enrichment and functional activity against leukemic blasts
- Consistent with their *ex vivo* alloreactivity, in two of the treated patients TK cells drove in vivo selection of immuno-resistant leukemic variants with genomic loss of the mismatched HLA haplotype

Figure 3. Thymic contribution to T cell recovery after infusion of TK cells

- After T cell-depleted haploidentical HSCT and the infusion of purified Tk^{pos} cells, reconstitution of Tk^{neg} CD3+ T cells occurs

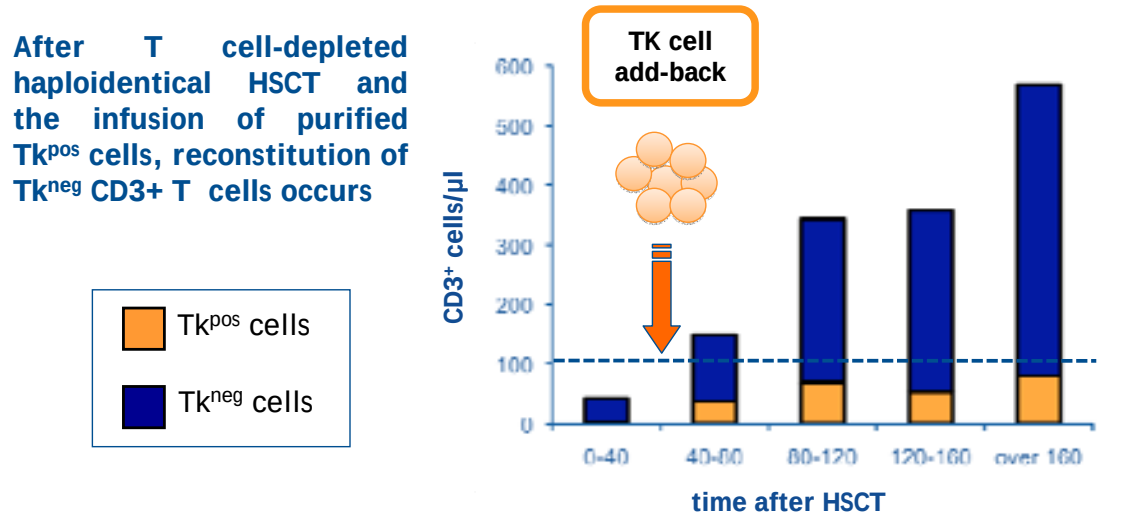

TK cell add-back
Tkpos cells
Tkneg cells
CD3+ cells/μl
0-40
40-80
80-120
120-160
over 160
time after HSCT

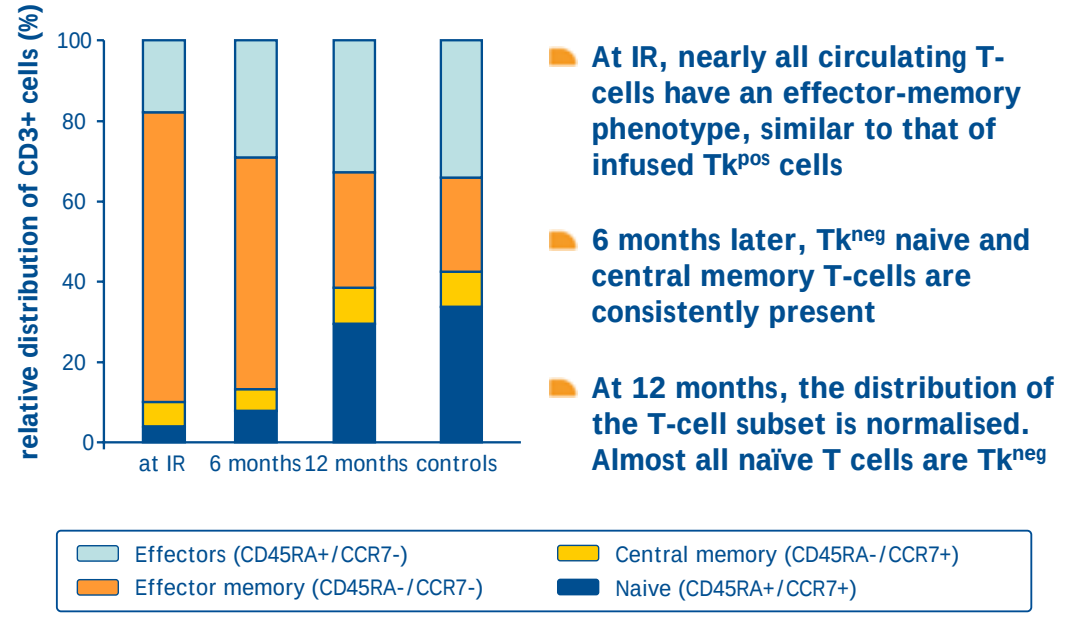

relative distribution of CD3+ cells (%)
at IR
6 months
12 months
controls
Effectors (CD45RA+/CCR7-)
Effector memory (CD45RA-/CCR7-)
Central memory (CD45RA-/CCR7+)
Naive (CD45RA+/CCR7+)

- At IR, nearly all circulating T-cells have an effector-memory phenotype, similar to that of infused Tk^{pos} cells
- 6 months later, Tk^{neg} naive and central memory T-cells are consistently present
- At 12 months, the distribution of the T-cell subset is normalised. Almost all naïve T cells are Tk^{neg}

- Naïve Tk^{neg} cells reconstituting after HSCT and Tk^{pos} cell infusion express PECAM-1 (CD31), a hallmark marker of Recent Thymic Emigrants (RTEs). Accordingly, an increase in thymic volume could be documented by CT scans.

Recent Thymic Emigrants (RTEs)

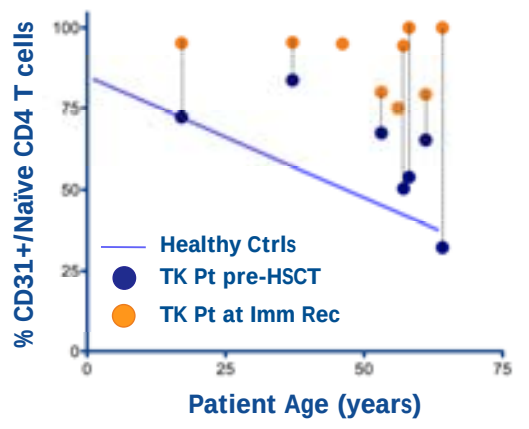

% CD31+/Naïve CD4 T cells
Healthy Ctrls
TK Pt pre-HSCT
TK Pt at Imm Rec
Patient Age (years)

Chest CT scans of UPN TK47 (age 64)

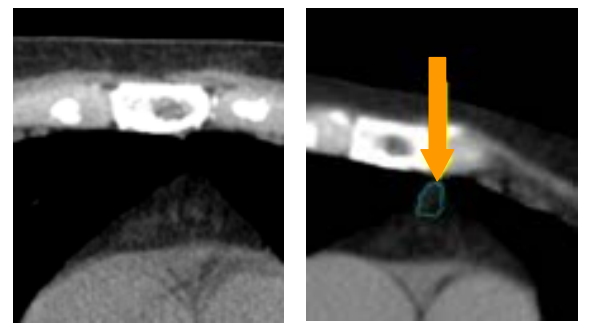

Pre HSCT
Thymic Index: 1

+60d post Tk-DLI
Thymic Index: 3

Figure 4. Protection against pathogens mediated by Tk^{neg} cells

- In case of GvHD, activation of the suicide gene by Ganciclovir (GCV) selectively eliminate Tk^{pos} cells, granting GvHD control in all treated patients

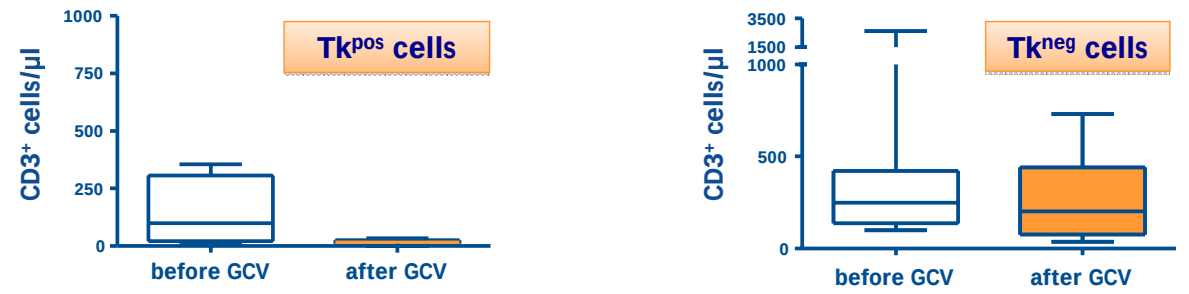

Tkpos cells
Tkneg cells
CD3+ cells/μl
before GCV
after GCV

- Elimination of Tk^{pos} cells by GCV did not hamper anti-infectious specific immunity nor increased subsequent infections, suggesting that pathogen control may be mediated by the newly generated thymus-derived Tk^{neg} cells

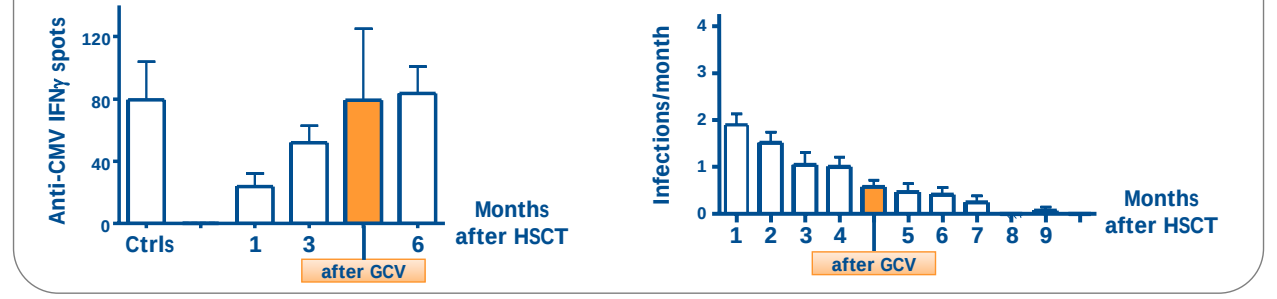

Anti-CMV IFNγ spots
Ctrls
Months after HSCT
after GCV
Infections/month
Months after HSCT
after GCV

Figure 5. Leukemia control through alloreactivity mediated by TK^{pos} cells

- In two of the patients treated with Tk cells, leukemia relapsed by losing the mismatched HLA haplotype targeted by donor T cells, suggesting that disease immune control strongly relies on Tk cell-mediated alloreactivity

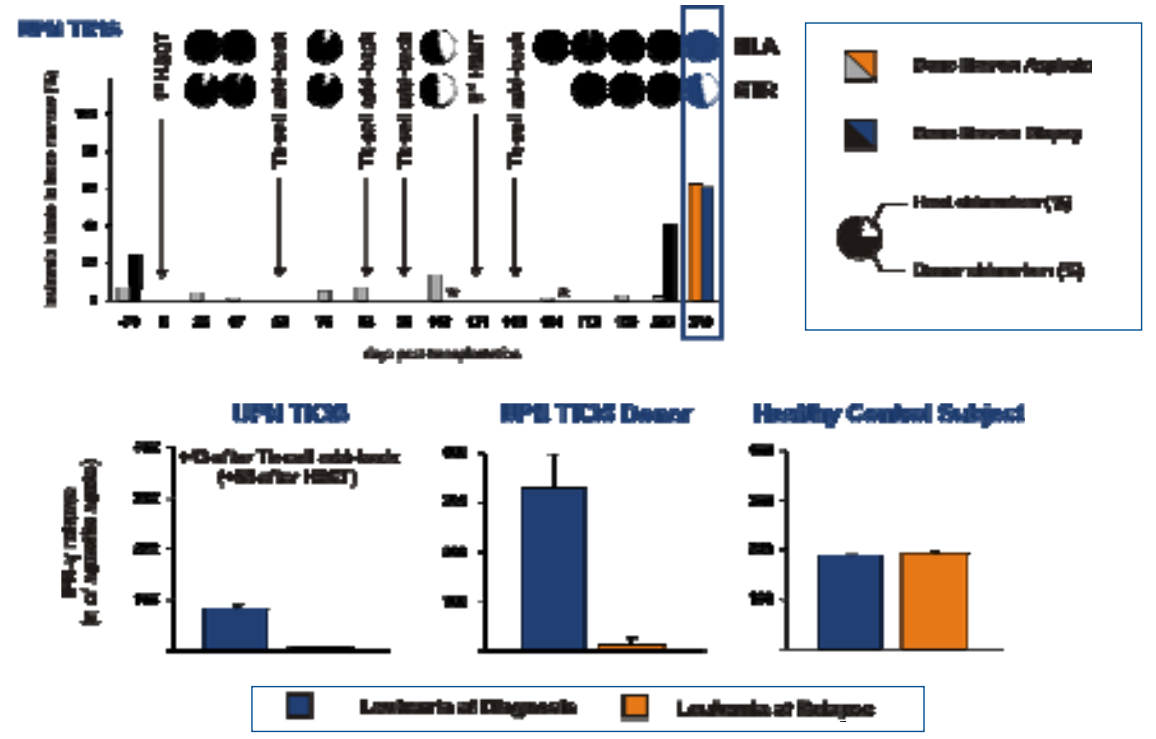

CONCLUSIONS

- The infusion of genetically modified donor T cells prompts renewal of thymic activity, which contributes to recovery of a polyclonal T cell repertoire
- Contextually, TK cells also mediate a direct anti-leukemic effect

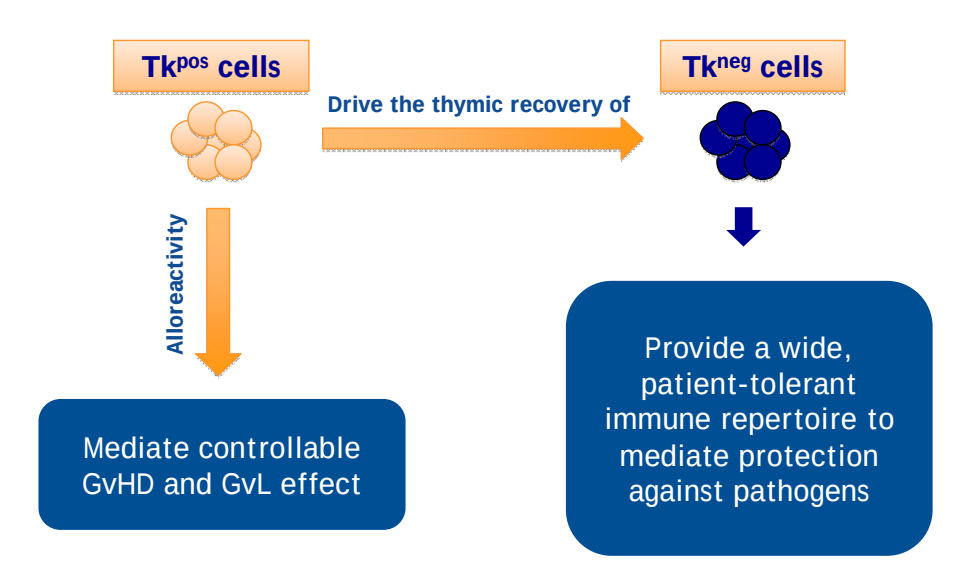

Tkpos cells
Drive the thymic recovery of
Tkneg cells
Alloreactivity
Mediate controllable GvHD and GvL effect
Provide a wide, patient-tolerant immune repertoire to mediate protection against pathogens

- TK cells activity in the context of haplo-HSCT is currently being assessed in a Phase III clinical trial (TK008)

References

1. Ciceri F. and Bonini C. et al. Lancet Oncol. 2009 10(5): 489-500
2. Kohler S. and Thiel A. Blood 2009 113(4): 769-74
3. Vago L. et al. N Engl J Med 2009 361(5):478-488

Acknowledgements

- Istituto San Raffaele: C. Soldati, S. Mastaglio, R. Greco
- MolMed: L. Callegaro, S. Colombi, A. Troysi, E. Lungagnani